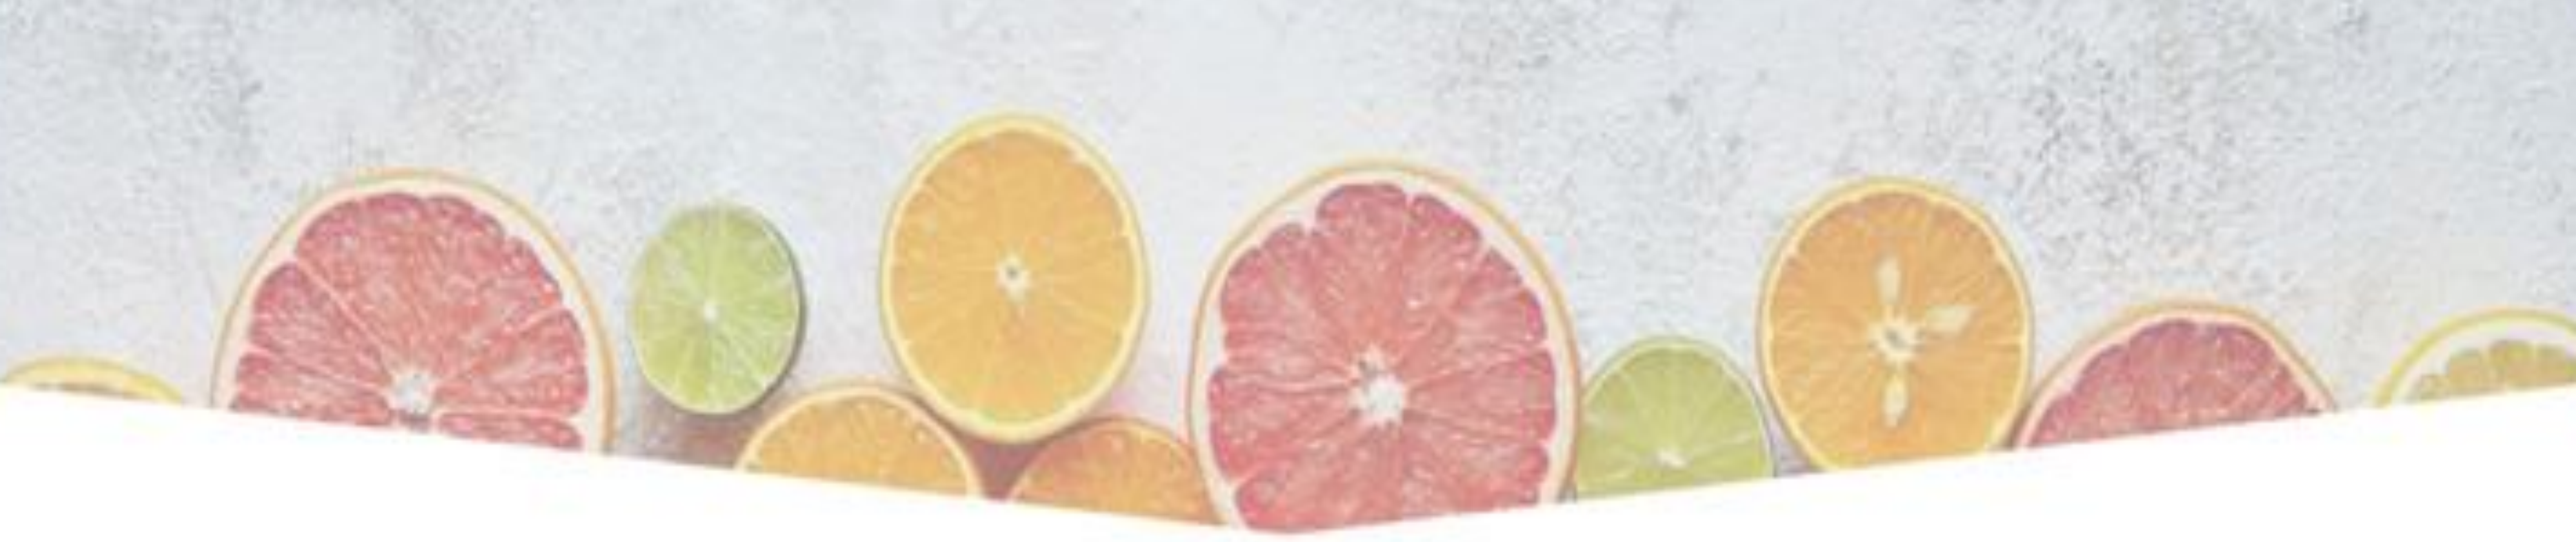

IPM-4-Citrus, Citrus disease

Integrated Pest Management: from Research to Market

Environmental toxicity assessment of NEW Bacillus thuringiensis kurstaki - based biopests on non-target organisms using ISO Standards and OECD guidelines.

Hazar KRAÏEM [1,2], Rim EL JENI [1], Gül AYYILDIZ [3] , Zakaria BENLASFAR [1], Zeynep YURTKURAN CETEREZ [3]
and Balkiss BOUHAOUALA-ZAHAR [1,4].

1 : Laboratoire des Venins et Molécules Thérapeutiques, Institut Pasteur de Tunis,13 Place Pasteur, BP74, 1002 Tunis Belvédère, Université Tunis-El Manar.2 : Laboratoires MédiS, Route de Tunis Km 7 - BP 206 Nabeul 8000 – Tunisie. 3 : Biyans Biyolojik Urunler Ar-Ge Dan. Tic. Ltd. Beycanoglu Is Merkezi no:104/20 Balgat Cankaya Ankara. 4 : Faculté de Médecine de Tunis, Université Tunis-El Manar.

Background and aim: Bacillus thuringiensis (Bt) was the first biopesticide widely used since 1957 against lepidopteron pests. The impacts on human and environment have to be evaluated, using toxicity assays according to the European guidelines. The aim of our research is to assess the biosafety of two new formulated Bt-based biopests on non-target organisms (i.e. Bt kurstaki BLB1 and LIP strains) by : (1) establishment of linear dependence of inhibition (I%) versus pesticide concentration, and (2) determination of the EC50 database for Bt-Lip concentrated growth culture.

Material and methods

Toxicity of B.t.k based biopest (LIP browth culture versus commercial formulation HD1-Delfin WG) on non-target organisms

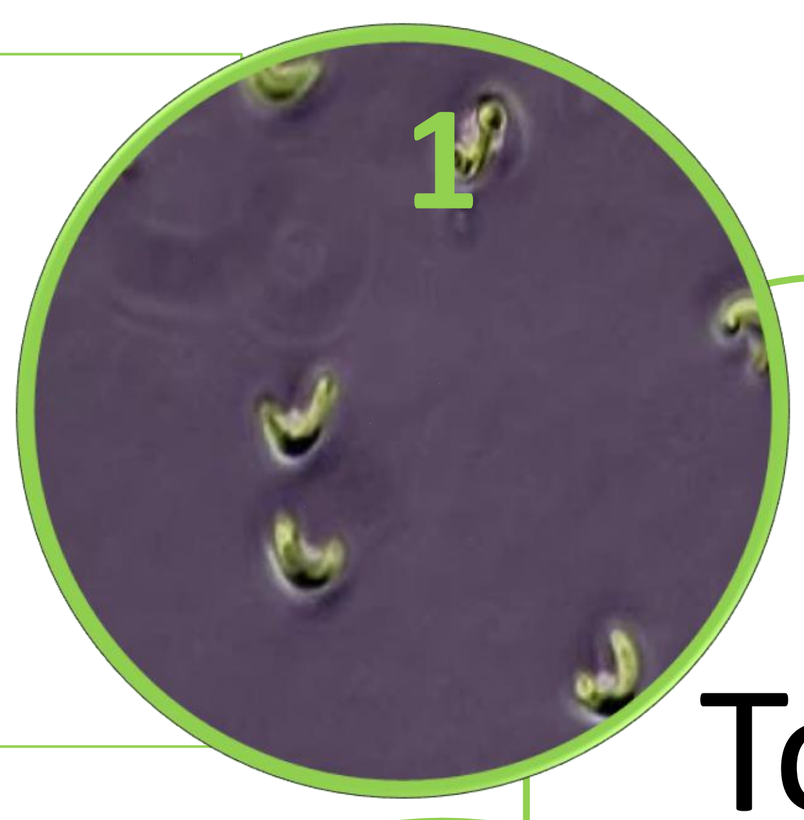

1

ALGALTOXKIT F FRESHWATER TOXICITY TEST WITH MICROALGAE

Selenastrum capricornutum (presently as Pseudokirchneriella subcapitata) .Measured concentration of 0, 6.25, 12.5, 25, 50, and 100 mg/L of δ-endotoxins→72h ErC50 deduced using Macro REGTOX.

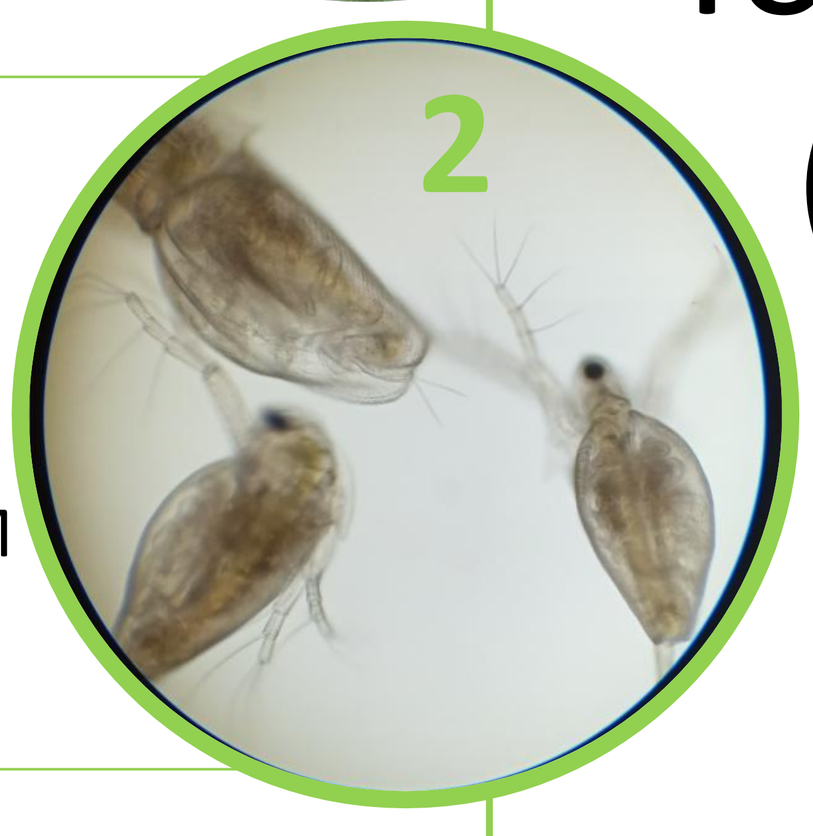

2

D A P H T O X K I T F TM MAGNA

Acute toxicity Test using **Newly hatched Daphnia magna larvae**, 3-day static renewal. Twenty individuals (4 replicates) were tested for each concentration : 0, 6.25, 12.5, 25, 50, and 100 mg/L of LIP vs Delfin WGTM (HD1) δ-endotoxins →48h ErC50 deduced using Macro REGTOX.

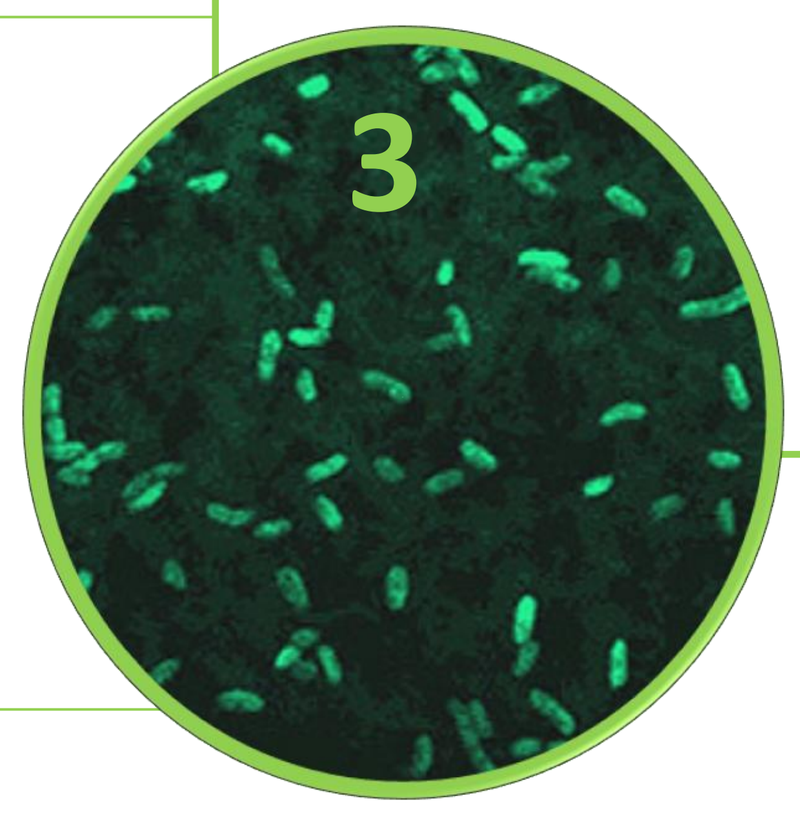

3

ALIIVIBRIO FISCHERI Bioluminescence Method

Toxicity Test using **Vibrio Fischeri luminescent bacteria .** All measurements were done on B.t.k LIP at 1000,500, 250, 125,62 and 31mg/L of LIP culture-δ-endotoxins. Bioluminescence measurements performed after 5 and 15 min exposure.

Results and Discussion:

Relationship between B.t.k -based biopesticide and percentage inhibition of Daphnia magna mobility in test lasting 24 and 48 hours

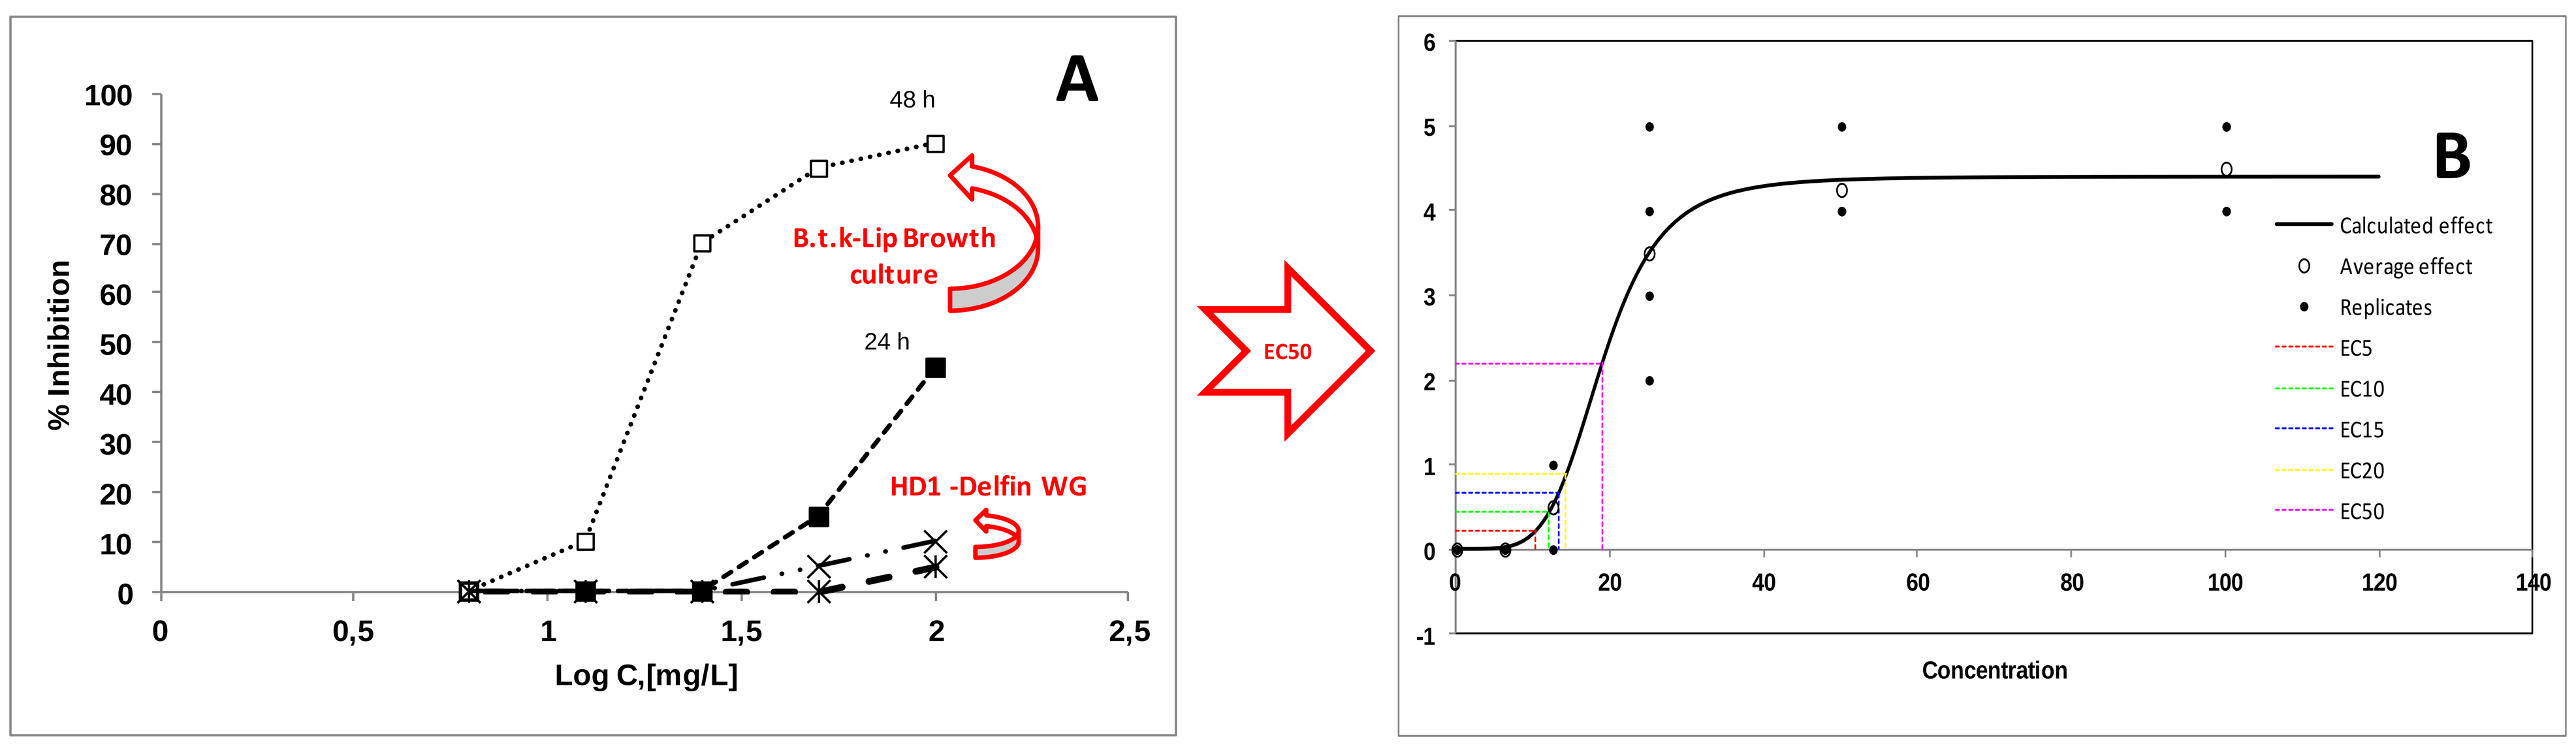

A
% Inhibition
Log C,[mg/L]
48 h
24 h
B.t.k-Lip Browth culture
HD1 -Delfin WG
EC50
B
Concentration
Calculated effect
Average effect
Replicates
EC5
EC10
EC15
EC20
EC50

❖**For D. Magna Acute toxicity test:**

✓The 48h ErC50 of Lip Growth culture: is estimated to **18,98 mg/L.**

✓The 48h ErC50 of HD-1 commercial formulation (Dipel TM) with is estimated to **68.01mg/L .**

✓the chronic daphnid no-observed effect concentrations , **NOEC is to 1,4 mg/L.**

⇨The Lip Growth culture is classified as **“moderately toxic”** to daphnids compared to HD-1 commercial formulation (Dipel TM) with a 21 day EC50 for immobilization of 14 mg/L and 10 d EC50 of 12 mg Bta/L for B. thuringiensis subsp. Aizawa (NOEC: 5.9 mg/ L (10-day)).

Lip toxicity effect on AliiVibrio Fischeri Bioluminescence after 5 and 15 min of exposure

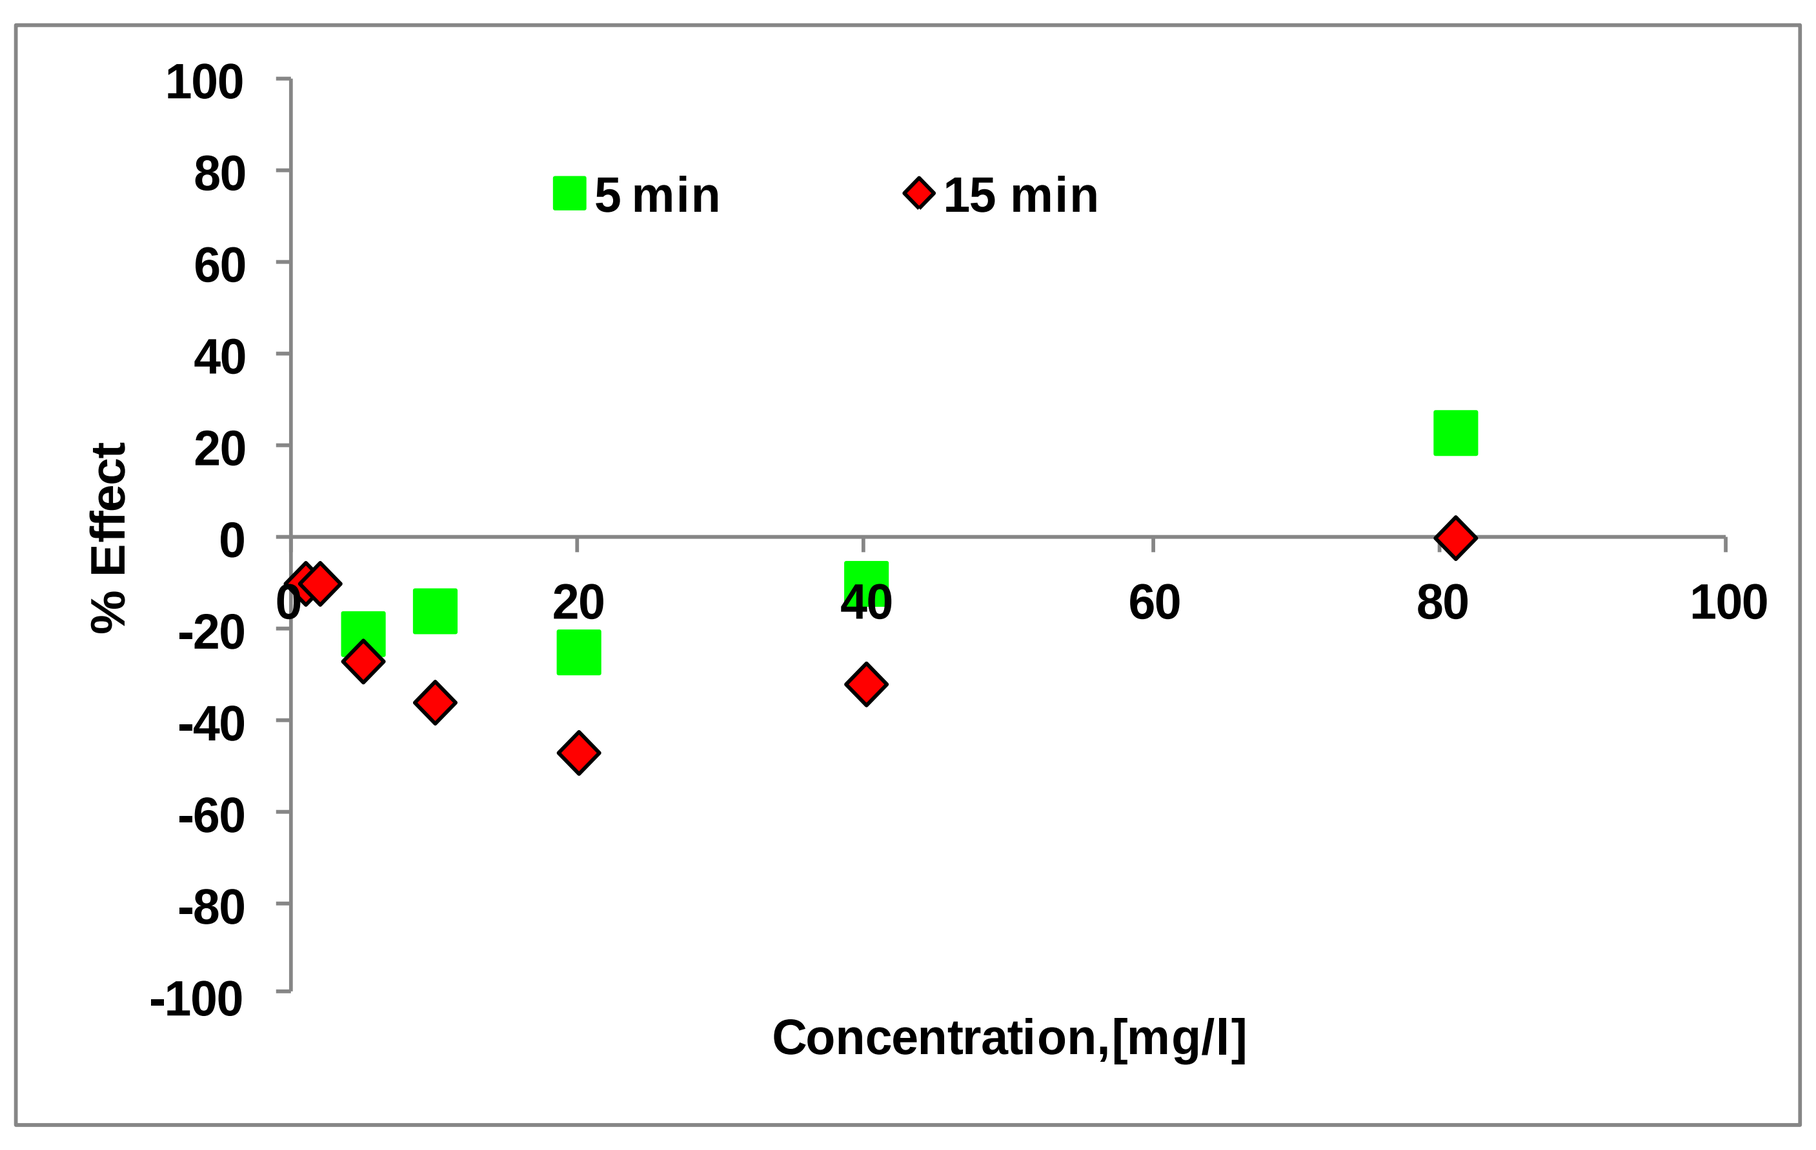

5 min
15 min
% Effect
Concentration,[mg/l]

❖**For Alii Vibrio Fischeri Toxicity test:**

Linear dependence between the Inhibition (%) and Lip-endotoxins concentrations but the test must be reproduced.

❖**For Freshwater, microalgae Toxicity test:**

The 72h Ec50 of Lip Growth culture is estimated to **81,80mg/L**

→ Higher than the EC50 values for B. thuringiensis subsp. Aizawai (72 h EC50 of 64.3 mg Bta/L; NOEC of 56.5 mg/L , Endpoint used for risk assessment.

Conclusion: Most organisms seem relatively tolerant even to concentrations of B.t.k. in water that are up to 200,000 times higher than expected environmental concentrations (Christensen 1991f). The results demonstrate that Cry1C protein has no lethal or sublethal effects on this non-target species.

This project has received funding from the European Union's Horizon 2020 research and innovation program under grant agreement No 734921.

IPM-4-CITRUS